Points to Consider for a Pharmaceutical Development "P2" Report for OINDP: An Industry View

Jackie Schumacher*, Alex Bell*, Mark Broughton, Mukul Dalvi, John Hart, Susan Holmes, Terry Kosobud, Paul Lucas, Lee Nagao†, Tilo Schönbrodt, Mary Ann Smith, Terrence Tougas, Tony Wilkinson, Xian-Ming Zeng

BACKGROUND

ICH M4 Guidance and subsequently Q8, written for section 3.2.P.2 Pharmaceutical Development, focuses on traditional pharmaceutical dosage forms.

INTRODUCTION

In 2007, the Model OINDP Working Group of the International Pharmaceutical Aerosol Consortium on Regulation and Science (IPAC-RS) developed and circulated to industry the Points to Consider for a Pharmaceutical Development Section for an Orally Inhaled or Nasal Drug Product. This document provided, for the first time, informal guidance based on industry best practices regarding the technical information that should be considered for inclusion in the P2 (Pharmaceutical Development) section of the Common Technical Document (CTD).

CHARACTERISTICS OF THE POINTS TO CONSIDER

The Points to Consider (PtC):

- Recognizes and encourages use of ICH Q8, Q9 and Q10 principles and quality by design concepts as they may be applied to aspects of OINDP development.
- Builds upon existing guidance provided by ICH Q8 adding specific recommendations unique to OINDP, including those related to:
 - Composition and properties of container closure systems and devices;
 - Aerodynamic particle size.
- Includes risk management approaches, based on current thinking from device development guidelines.
- Is a "living document," and will be revised periodically using feedback from industry and regulators. In early 2008, the document was submitted to the FDA for comment.

DISCUSSION

Due to the route of administration, particular attention has been given to factors that may affect delivery to the lungs or nasal passages, such as the container closure system/device; therefore, this section of the Points to Consider document has been developed in more detail.

Device development is an iterative process: the user needs, or intended use, are first defined and then translated into specifications, which become the design inputs and are used to develop the device design. This design process should result in a prototype, drawing, or blueprint of the device, which is then verified to ensure that the design inputs, originally specified and validated against the defined user needs, are met. If the design inputs or user needs are not met, the design process, followed by verification and validation, is repeated until the design output fulfills all requirements, at which point the product is suitable for commercial manufacture.

Figure 1. depicts design controls, which are typically used in device development, and the role of risk analysis in this process. This figure may be helpful in understanding the container closure system/device design process.

Risk analysis is a key element in every step of this process; this approach helps to define the user needs, identify and manage potential areas of risk, and identify key quality attributes.

Figure 1. Device Design Controls (Adapted)

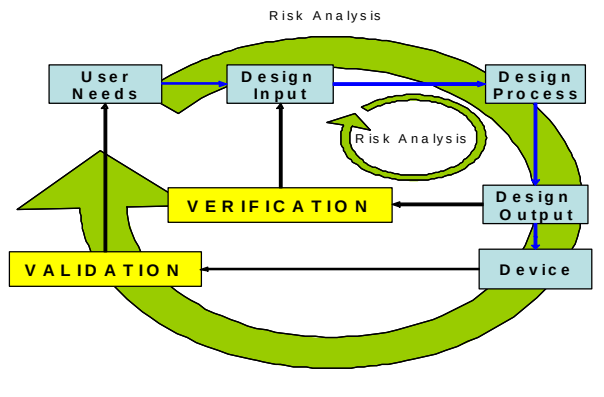

Risk Analysis
User Needs
Design Input
Design Process
Risk Analysis
VERIFICATION
Design Output
VALIDATION
Device

Adapted from Figure 1, FDA Guidance for Industry: Design Controls for Medical Device Manufacturers, CDRH, March 1997, and originally used with permission of the Medical Devices Bureau, Health Canada. FDA Guidance endorsed by the Global Harmonization Task Force June 1999. Available at: http://www.fda.gov/cdrh/comp/designgd.html

Importance of the Container Closure System.

As indicated above, the container closure is of primary importance to OINDPs. Accordingly, 3.2.P.2.4 Container Closure System should follow a logical organization; e.g., an MDI with a canister, valve, actuator, over-wrap, etc., may be presented in one section (container closure system), whereas a pre-dispensed unit with a separate mechanical device may be presented in separate sections (container closure system and device). Because the formulation and the container closure system/device in an OINDP frequently interact in a significant way to affect device or product performance, the formulation and final drug product are discussed where relevant.

Per ICH Q8, sufficient information and detail should be included in this section to provide a comprehensive understanding of the container closure system, device, and their development and interaction with the formulation.

Table 1. Proposed Sections within 3.2.P.2.4

Proposed Section Title	Content
Intended Use	This section might describe the intended use for the container closure system/device and provide the rationale for selection of the design input requirements, e.g., by addressing: • intended use of the final drug product, and • an acceptable risk profile
Risk Management	The philosophy governing the approach to risk analysis used in the container closure system/device development program may be described here. Specific risk analyses and their results may be discussed in the sub-sections below as appropriate to justify decisions made, and cross-referenced here. The level of detail in these discussions will vary depending on the complexity of the container closure system/device and the frequency and severity of the risks.
Prior Knowledge	This section could describe any relevant information from container closure systems used previously, for example, from platform systems/devices.
Design Inputs	This section might describe the detailed design input requirements for the container closure system/device components together with a rationale for their inclusion. These may be derived from: • the intended use, risk analysis, and prior knowledge described in the previous sections • compatibility and interaction of the container closure system/device with the formulation, and • the relevant regulations and standards. Justification of any deviation from the relevant regulations and standards should be considered for inclusion in this section.
Design Outputs	This section might include a general description of the container closure system/device and describe how the container closure system/device meets the design input and protects against identified risks.
Design Verification	This section may demonstrate that the design input requirements are fulfilled by the design of the container closure system/device. The following may be considered: • a general overview of the design verification strategy, • verification methods, results and conclusions

Table 1. (cont'd) Proposed Sections within 3.2.P.2.4

Proposed Section Title	Content
Design Validation	This section may describe studies to demonstrate that container closure system/device requirements conform to the user needs and intended use described previously. This section may include: • description of the techniques employed to validate the design, e.g., mechanical, functional, and/or clinical studies, the rationale for their selection, and results. • any risk analyses conducted to support the rationale and/or choice of validation techniques. • drug product characterization studies used to demonstrate that the container closure system/device fulfills user needs and/or to support label instructions and the rationale for their selection and inclusion
Manufacturing Process	This section, with reference to 3.2.P.2.3 Manufacturing Process Development, might describe the manufacture of the container closure system/device and the development of that manufacturing process. This section may include: • a description of the approach to the development of the manufacturing process including major design inputs for the manufacturing process e.g., critical quality attributes, risk analyses, production specifications, etc. • a discussion of the final assembly process for the container closure system/device and how the container closure system/device assembly process and the drug product manufacturing process interact (e.g., during filling) • a discussion of any design or manufacturing process changes made during scale-up from initial to final commercial design. These may be discussed in terms of their impact on the product's quality attributes. As an example, a risk assessment for advancing from single to multiple cavity tooling may be included. • validation studies conducted to demonstrate the ability of the manufacturing process to reliably produce a container closure system/device with the appropriate quality.

Considering the characteristics of device design controls and the proposed elements of the Container Closure section, the working group pursued the opportunity for further applying Quality by Design principles. In Figures 2-5, the relationships among the product profile and the specific technical requirements for OINDP are illustrated in order to clarify the guiding philosophy and approaches expressed in the Points to Consider.

Product Lifecycle.

Figure 2 provides an overall product lifecycle flowchart based on risk management, starting with establishment of a product profile, and linking subsequent steps to the profile and technical requirements needed to fulfill that profile.

Figure 2. Product Lifecycle Flowchart.

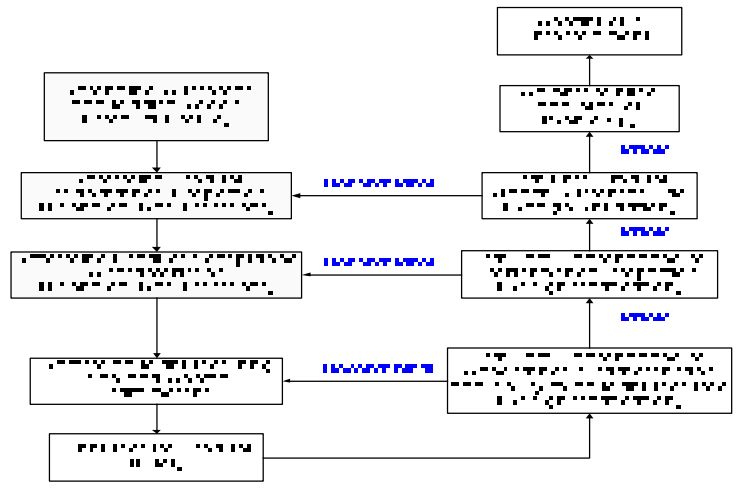

It is recognized that the relationship between particle size and clinical outcomes, e.g., user and technical requirements - such as particle size distribution (PSD), has not been established in most cases. However, in cases where this relationship can be inferred, this discussion might include:

a) describing the rationale for the studies (in vivo/in vitro) performed to evaluate the effect of the particle size distribution as it relates to the desired product profile;

b) identifying the site of deposition of different particle sizes (e.g., in the oropharynx, large airways, smaller airways, and respirable dose);

c) establishing a correlation or relationship between the range of particle size distributions studied and the clinical outcomes; and finally,

d) justifying the acceptance criteria, if applicable, for particle size distribution based on the clinical relevance.

Product Profile and Technical Requirements Specification.

Figure 3 shows an example product profile and high-level technical requirements for an OINDP, in this case, the elaboration pertains to an MDI. The information is obtained through the interactions of a multidisciplinary team of individuals, with focus on the end product attributes. Over the course of development, i.e. as technical data are gleaned, the relevance of certain attributes may change. However, the concepts associated with Device Design Controls, Figure 1, continue to be applied.

Figure 3. Example Product Profile and Technical Requirements for an MDI

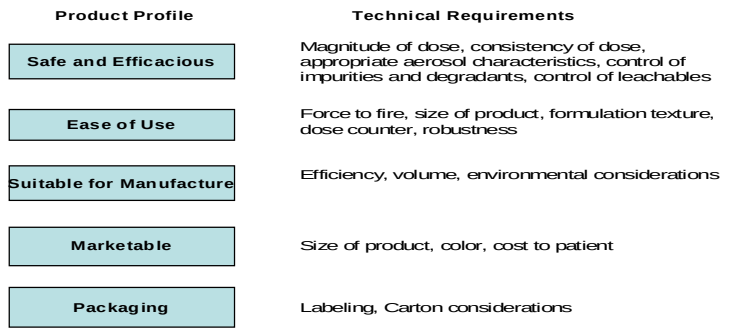

Product Profile
Technical Requirements
Safe and Efficacious
Magnitude of dose, consistency of dose, appropriate aerosol characteristics, control of impurities and degradants, control of leachables
Ease of Use
Force to fire, size of product, formulation texture, dose counter, robustness
Suitable for Manufacture
Efficiency, volume, environmental considerations
Marketable
Size of product, color, cost to patient
Packaging
Labeling, Carton considerations

Technical Requirement Linkages to Target Product Profile.

Figure 4 shows how specific technical requirements can be linked to the product profile, using PSD as an example. Key technical requirements critical to fulfilling the product profile can be identified in this way.

Figure 4. Technical requirements associated with product profile, using PSD as an example. Elements bound in red are linked to Examples.

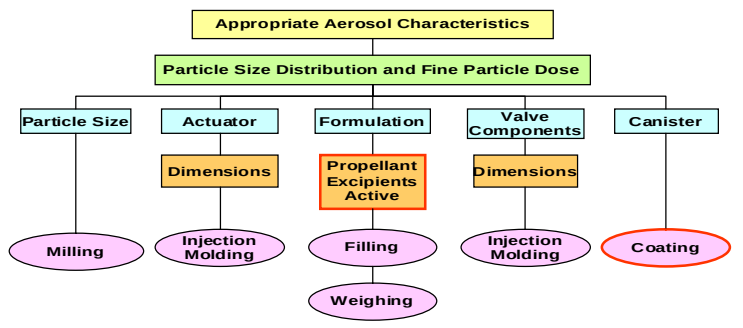

Appropriate Aerosol Characteristics
Particle Size Distribution and Fine Particle Dose
Particle Size
Actuator
Formulation
Valve Components
Canister
Dimensions
Propellant Excipients Active
Dimensions
Milling
Injection Molding
Filling
Injection Molding
Coating
Weighing

Examples.

As noted in Figure 4, examples of information and justifications related to specific technical requirements that could be included in a P2.

Canister Coatings

Coatings may inhibit crystallization of API, which may affect particle size distribution. Describe the intended use and need for the specific coating. Work with the supplier to understand the coating process and coat ingredients. Describe risks to particle size distribution associated with use of coating. Show how final product fulfills design inputs and meets intended use/user needs.

Formulation Excipients

Ostwald ripening is a well-known phenomenon that could occur in OINDP suspension formulations, and which can significantly affect the stability of the PSD attribute, since final particle sizes would be determined by this process. Describe the intended purpose of the excipients used. Explain the possible effect of excipients used and influence of temperature on Ostwald ripening for the specific formulation. Describe risks to PSD and link final outcomes to design inputs and intended use/user needs.

CONCLUSIONS

The working group hopes that this work product will be utilized by industry. Feedback on this edition of the PtC will provide the direction for future amendments and help to refine industry best practices. The working group believes that legacy MDI examples, cited here, will help to pave the path for innovative/novel OINDP systems.

NEXT STEPS

Next steps for the Model OINDP Working Group efforts include:

1) Continuing to gather industry OINDP development experiences

2) Cataloguing and incorporating feedback into subsequent revisions of the Points to Consider.

In addition, IPAC-RS plans to further progress its understanding of the relationship between quality attributes, e.g. particle size and clinical outcomes (Quality/Clinical Correlations Working Group).

ACKNOWLEDGEMENTS

The authors of the poster would like to gratefully acknowledge Liuda Shtohryn from AstraZeneca, previous Co-Chair of the Model OINDP WG, along with IPAC-RS colleagues who previously served on the working group and contributed to the Points to Consider document: Claire D'Abreu Hayling, Teva; Stefan Leiner, Boehringer Ingelheim; Michael Golden, Pearl Therapeutics; Steve Nichols, formerly with sanofi-aventis; Laurence Huxham, Novartis; and Johan Waldeck, Novo Nordisk. Melinda Munos formerly from the IPAC-RS secretariat was a significant driver for this effort and we commend her diligence and tenacity.

In addition to the industry representation, the Working Group would like to thank ONDQA pulmonary chemists for their willingness to review the PtC and their subsequent comments, now incorporated into the PtC.

*Co-Chairs of the Working Group †Corresponding Author (lee.nagao@dbr.com); Schumacher, Hart, Lucas (Pfizer); Bell, Wilkinson (3M); Broughton (sanofi-aventis); Dalvi (Novartis); Holmes (GSK); Kosobud (Abbott); Schönbrodt, Tougas (Boehringer Ingelheim); Smith (Nektar); Zeng (Teva)