A "Compare and Contrast" of International Guidelines Impacting the Pharmaceutical and Clinical Development of New and Subsequent Market Entry OINDPs Part A: Clinical

Jackie Schumacher*, Ray Ormiston*, Bob Alekel, Bettina Berner, Nola Bowles, Mike James, Jim Keegstra, Lana Lyapustina†, Andy Rignall, Cheryl Rogerson, Brett Thompson

BACKGROUND

Over the last several years, a number of national and international standard-setting bodies and regulatory agencies have issued a variety of final and draft guidelines pertaining to the pharmaceutical and clinical development of orally inhaled and nasal drug products (OINDPs). Some of these requirements are similar across guidelines and regions, while some are less harmonized.

During 2005, significant international focus was placed on the development of OINDP with the publication of the joint draft guidance by Health Canada and European Medicines Agency/CHMP (Quality Working Party). At that time, IPAC-RS solicited member company feedback and comments for transmittal to both regulatory agencies. Based on the substantial comments, the Quality Working Party and Health Canada determined the necessity to convene an Interested Parties meeting. IPAC-RS collaborated with EfPIA and other interested parties to provide a consolidated industry view and perspective during the discussion with the QWP. Participation in these activities prompted IPAC-RS to prioritize the importance of maintaining an active international focus.

INTRODUCTION

Since 2005, the International Industry Coordinating Group (IICG) has identified opportunities to reach out, including sponsoring a roundtable regulatory discussion with representatives from EU and Canadian national agencies affiliated with the now final orally inhaled and nasal pharmaceutical guideline In addition, the IICG has reviewed and provided consolidated industry comments on international regulatory guidelines pertinent to the clinical and pharmaceutical development of OINDP, both new and subsequent market entry (generic). These are listed under 'Representative OINDP Specific Medical Guidelines,' indicated by an asterisk. The clinical requirements of these guidelines have been reviewed and compared and contrasted below. The IICG hope to stimulate discussion between industry, academia, and the regulators on areas of interest requiring further development, and some challenges are listed below.

DISCUSSION

The cost of medicines is an important factor for patients and the healthcare industry as a whole. Significant emphasis in the development of second entry or "generic" medicines for patients with asthma, COPD and other lung diseases has been a topic of recent advisory discussions and guideline development worldwide.

There is general recognition in the three subject regions, i.e. US, EU, Canada, regarding the current limitations for establishing an in vivo/in vitro correlation (IVIVC) in these medicines, e.g. locally acting OINDP. Therefore, sponsors and applicants are taking a variety of approaches to demonstrate equivalence or comparability. In addition, there is specific focus on the use of relevant biomarkers which may facilitate the study of these medicines. These points will be briefly discussed here.

REGULATORY FRAMEWORK AND LANDSCAPE

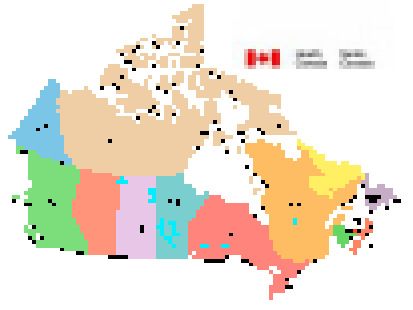

http://wwp.greenwichmeantime.com/time-zone/north-america/canada/_derived/index.htm_txt_canada-map.gif

Health Canada (HC).

- § Subsequent market entry ICS products that are considered to be new drugs are subject to the requirements of Division 8, Part C of the Food and Drug Regulations. Abbreviated New Drug Submissions (ANDS) and Supplemental New Drug Submissions (SNDS) in support of these drug products are required to contain evidence of safety and effectiveness under the proposed conditions of use, in conformity with the applicable sections of the Food and Drug Regulations namely: C.08.002.1, C.08.003 and C.08.005.1 (**http://laws.justice.gc.ca/PDF/CRC_870.pdf**)
- § According to the HC Draft Guideline: "Clinical Requirements for Second-Entry Inhaled Corticosteroids for the use in the Treatment of Asthma " (2007) the generic product application must include at least: a) PK comparison for safety; b) PD comparison for efficacy - Parallel or cross-over, double-blind/Relative potency: more than one dose
- § Relevant biomarkers – airway eosinophils and exhaled nitric oxide (eNO), where eosinophils were advocated by Canadian advisory at RDD 2008 (Irwin Mayers, University of Alberta).

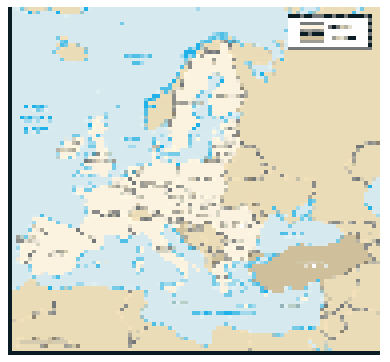

http://wwp.greenwichmeantime.com/time-zone/europe/european-union/images/map-of-eu.gif

European Medicines Agency (EMEA).

- § Directive 2001/83/EC (as amended) of the European Parliament defines the requirements for medicinal and generic products, specifically, Article 8 for medicinal product and Article 10 for generic products; **http://www.emea.europa.eu/pdfs/human/pmf/2001-83-EC.pdf**
- § The concept of therapeutic equivalence for OIP is precedented with generic inhaled products available on the EU market.
- § PtC on Clinical Requirement for OIP – a) Product similar to an existing one - in vitro data, inhalation device is identical, differences in excipients known to have no influence; b) Product not similar to an existing one - Clinical trials / PD studies
- § Relevant biomarkers – biomarkers with sufficient sensitivity, no preference identified.
- § Comparative APSD studies should be based on statistically relevant observations. IPAC-RS has commented that the EU OIP guideline may not be suitable, as currently drafted.
- § In the EU draft OIP guideline, recommendations for demonstration of therapeutic equivalence for ICS are quite challenging, especially in light of the lack of clinical assay sensitivity for ICS dose-response. Moreover, the recommended approaches e.g., the selection of patients that have demonstrated an ICS dose-response at screening; and the need to demonstrate dose-response in the same study for different strengths of LABA/ICS combinations have no precedent in the literature. These CHMP proposals may, therefore, be premature.

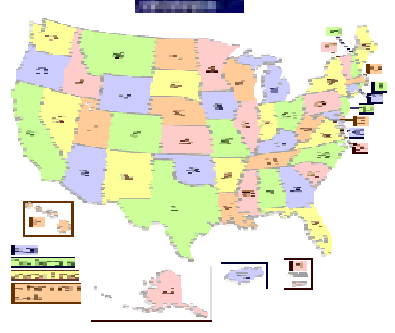

http://www.epa.gov/OGWDW/ccr/us-regions_files/us-states.gif

US Food and Drug Administration (FDA).

- § Three types of applications exist: 1) full safety, efficacy, and relevant data conducted by applicant, i.e. Section **505(b)(1)** of the FDC Act; 2) safety and efficacy by applicant, other data may not have been obtained by right of reference, i.e. Section **505(b)(2)**; 3) contains information to show that the proposed product is identical in active ingredients, dosage form, strength, route of administration, labeling, quality, performance characteristics, and intended use, to a previously approved product, i.e. **Section 505(j)**.
- § According to the document "Critical Path Opportunities for Generic Drugs" (1st May 2007) the generic product application must include: a) In vitro comparison; b) PK comparison for safety; c) PD comparison for efficacy - Relative potency
- § Approval of generic drugs in the United States requires demonstration of BE of the generic drug; to the reference listed drug (RLD), therefore there are limited generic OINDPs on the US market, e.g. albuterol.
- § No established models for demonstration of BE of ICS due to lack of clinical models showing dose response relationships. The only available guidance is the 1994 PtC detailing "switch" program.
- § "Asthma stability model" – work in progress and showing promise – Ahrens (RDD 2007); subject of Pharmaceutical Sciences Advisory Committee questions (July 2008)
- § Relevant biomarkers – exhaled nitric oxide (eNO) shows promise and research is underway.

Representative OINDP Specific Medical Guidelines.

As noted earlier, the IICG has created a compendium of relevant guidelines to the development of OINDP. A snapshot of OINDP specific medical guidelines follows:

Canada

Health Canada (2007) Draft Guidance Document: Submission Requirements for Subsequent Market Entry Inhaled Corticosteroid Products for Use in the Treatment of Asthma*
http://www.hc-sc.gc.ca/dhp-mps/prodpharma/applic-demande/guide-ld/index_e.html

Health Canada (2007) Draft Guidance Document Submission Requirements for Subsequent Market Entry Steroid Nasal Products for Use in the Treatment of Allergic Rhinitis*
http://www.hc-sc.gc.ca/dhp-mps/prodpharma/applic-demande/guide-ld/index_e.html

EMEA

CPMP/EWP/4151/00 Rev. 1 (Draft Guideline on the requirements for clinical documentation for orally inhaled products (OIP) including the requirements for demonstration of therapeutic equivalence between two inhaled products for use in the treatment of asthma and chronic obstructive pulmonary disease (COPD)*
http://www.emea.europa.eu/pdfs/human/ewp/415100enrev1.pdf
and Appendix 1EMEA/CHMP/EWP/48501/2008*
http://www.emea.europa.eu/pdfs/human/ewp/4850108en.pdf

CPMP/EWP/2922/01 Note for guidance on the clinical investigation of medicinal products in the treatment of asthma.
http://www.emea.europa.eu/pdfs/human/ewp/292201en.pdf

US

Point to Consider. Clinical Development Programs for MDI and DPI Products (Sept 1994)
http://www.fda.gov/CDER/GUIDANCE/old046fn.pdf

Draft Guidance Chronic Obstructive Pulmonary Disease: Developing Drugs for Treatment (2007)
http://www.fda.gov/cder/guidance/7875dft.pdf

Draft Guidance for Industry Sinusitis: Designing Clinical Development Programs of Non-antimicrobial Drugs for Treatment (2006)
http://www.fda.gov/cder/guidance/7316dft.pdf

Challenges in Clinical Development.

- § Bioequivalence criteria, in comparison to existing BE guidance
- § Consistent requirement for dose response in ICS's
- § Extrapolation of data from asthmatics to patients with COPD
- § Higher strengths OINDP
 - § PD study only possible with the lowest strength
 - § Proportional lung deposition
- § Combination products
 - § In vitro attributes, characteristics for comparison
 - § Lung deposition studies
 - § PK safety
- § Lung deposition studies - Imaging studies
 - § Scintigraphy
 - § PK studies: with or without active charcoal

Questions and Considerations.

Are in vivo/in vitro correlations possible for OINDPs – for locally acting, or for systemically delivered? That is, can a clear Quality, PK/PD relationship be established?

Can differences in product quality be detected with primary clinical endpoints?

Typically PK studies are used to assess systemic safety. If systemic concentrations are not measurable, then a pharmacodynamic endpoint may need to be considered to assess safety. Do all regions agree with this view?

For "generic" OINDPs, there appears to be an opportunity to harmonize globally in vitro equivalence/similarity. What efforts can industry undertake to progress this concept, e.g. COPD guideline update for HC subsequent market entry products?

Also for "generic" OINDPs, what is the best method to determine deposition similarity, e.g. imaging studies?

In the US, the recent Pharmaceutical Advisory Committee reviewed potential models for OINDP clinical development. Clearly the direction of this activity will be an important factor for OINDP manufacturers.

What biomarkers, if any, have shown suitable sensitivity for OINDP?

Would an industry-generated risk-based decision tree be helpful for regulatory agencies?

When do HC and EMEA anticipate the draft guidelines will become final? Is there any opportunity harmonize the expectations between the two regions similar to the Pharmaceutical Guideline for OIP? And will there be any opportunity for discussion with Interested Parties, based on the existing comments?

FUTURE CONSIDERATIONS

Next steps for the IICG efforts include:

- Supporting the PQRI/INTFG BE Workshop in 1Q2009.
- Gaining a better understanding of the recent publications of Chinese and Brazilian OINDP guidelines/requirements, as these apply to both clinical and CMC considerations.
- Continuing discussions with clinicians, academicians and other industry representatives to further progress a better understanding of the possibilities of IVIVCs for OINDPs.
- Fostering the collection of data to substantiate or progress the clinical concepts advocated in the three regions.

ACKNOWLEDGEMENTS

The authoring team would like to thank colleagues, Sharon Gorman (Pfizer), Paul Lucas (Pfizer), Bob Berger (Schering Plough), David Cipolla (Aradigm) and Terry Tougas (BI) for stimulating discussions on these topics.

The working group would also like to acknowledge the support of the Day 3 CMC/Clinical panelists for the IPAC-RS 2008 Conference, for developing key thoughts and ideas for discussion and debate.

*Co-Chairs of the Working Group †Corresponding Author (**svetlana.lyapustina@dbr.com)** Schumacher (Pfizer); Ormiston, James (GSK); Alekel (Schering-Plough), Berner (Boehringer Ingelheim), Bowles (3M), Keegstra (Teva), Rignall (AstraZeneca), Rogerson (Novartis), Thompson (sanofi-aventis)