Analytical methods Quality by Design (AM-QbD) Risk Assessments undertaken within GlaxoSmithKline (World Wide Inhaled Product Development) for Orally Inhaled and Nasal Products

Andrew J Crumpton, Geoffrey E Daniels, Karl M Ennis and Nick C Turner. With Acknowledgements to Andrew J Rice.
GlaxoSmithKline R&D, Ware, Hertfordshire, UK

Introduction

Fundamental to any method development is being clear on the design intent of the method. **Method Performance Criteria** and **Method Operational Intent** are two important aspects of this design intent.

Project Background

Throughout the lifecycle of a given Orally Inhaled and Nasal Drug Product (OINDP) method it is inevitable that there will be changes in the 'method environment' that can impact its operation. Changes/improvements to a method should be made with reference to a method knowledge repository. GSK's repository is entitled **EMPACT** (**Experimental Method Parameter Assessment Control Tool**).

Aims

Ø Describe the repository and provide an overview of the analytical method risk assessment process used at GSK for OINDPs with some examples for MDI and DPI Products.

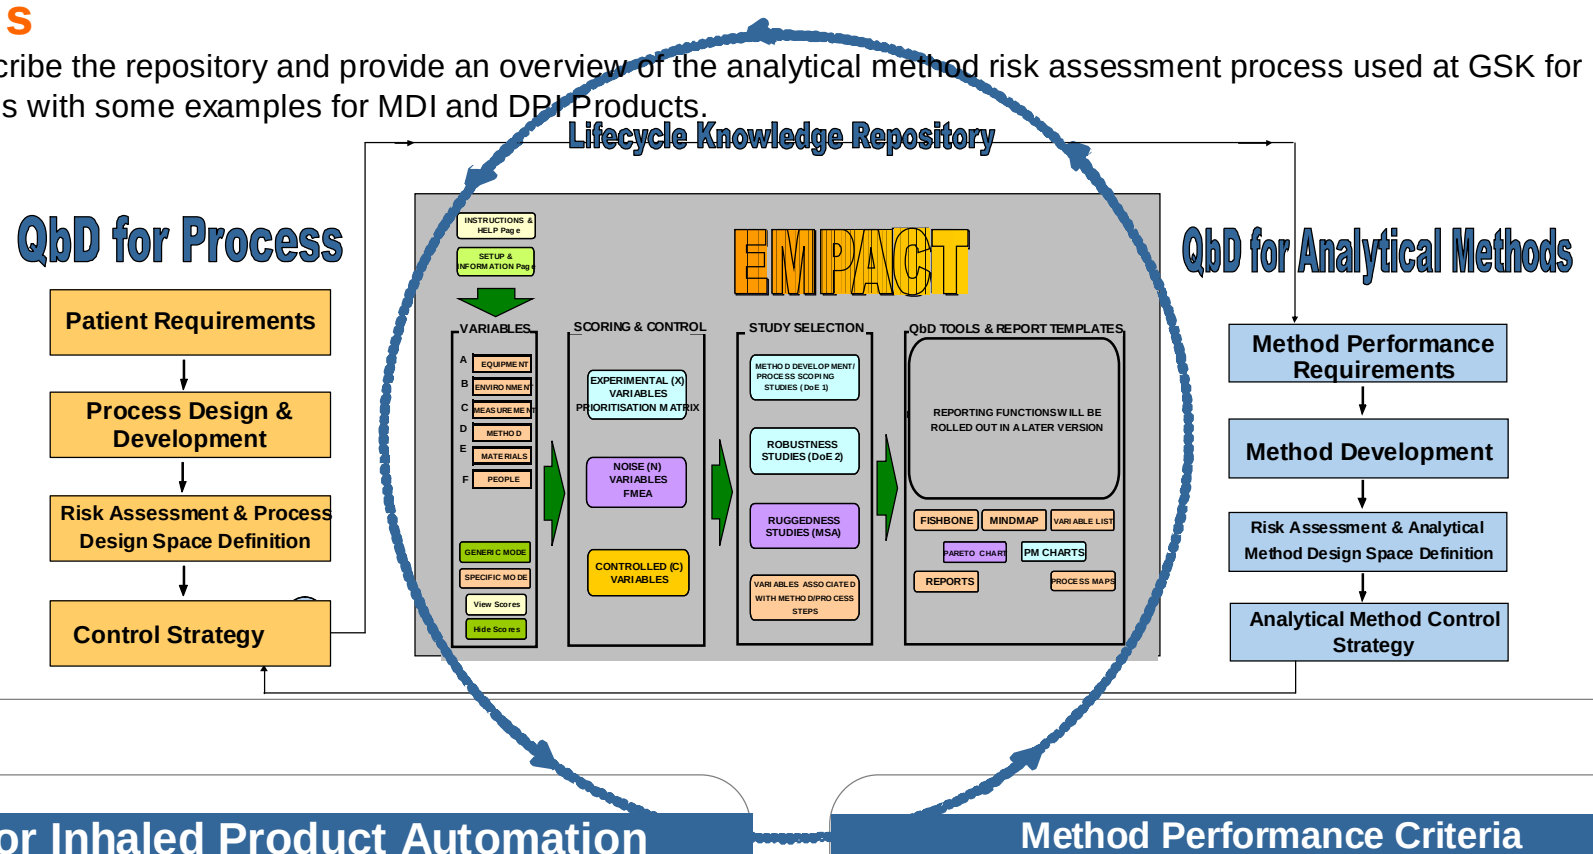
Lifecycle Knowledge Repository
QbD for Process
Patient Requirements
Process Design & Development
Risk Assessment & Process Design Space Definition
Control Strategy
EMPACT
INSTRUCTIONS & HELP Page
SETUP & INFORMATION Page
VARIABLES
A EQUIPMENT
B ENVIRONMENT
C MEASUREMENT
D METHOD
E MATERIALS
F PEOPLE
GENERIC MODE
SPECIFIC MODE
View Scores
Hide Scores
SCORING & CONTROL
EXPERIMENTAL (X) VARIABLES PRIORITISATION MATRIX
NOISE (N) VARIABLES FMEA
CONTROLLED (C) VARIABLES
STUDY SELECTION
METHOD DEVELOPMENT/ PROCESS SCOPING STUDIES (DoE 1)
ROBUSTNESS STUDIES (DoE 2)
RUGGEDNESS STUDIES (MSA)
VARIABLES ASSOCIATED WITH METHOD/PROCESS STEPS
QbD TOOLS & REPORT TEMPLATES
REPORTING FUNCTIONS WILL BE ROLLED OUT IN A LATER VERSION
FISHBONE
MINDMAP
VARIABLE LIST
PARETO CHART
PM CHARTS
REPORTS
PROCESS MAPS
QbD for Analytical Methods
Method Performance Requirements
Method Development
Risk Assessment & Analytical Method Design Space Definition
Analytical Method Control Strategy

Process Map linked to DFM

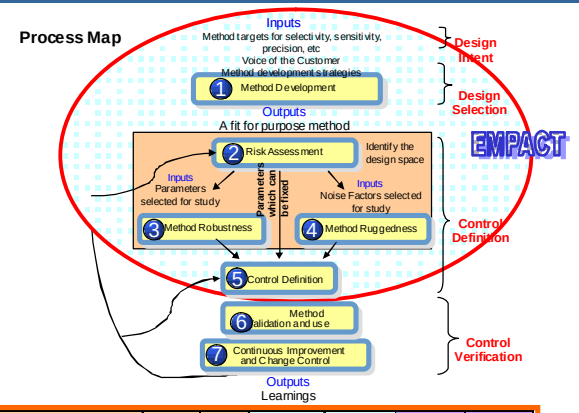
Process Map
Inputs
Method targets for selectivity, sensitivity, precision, etc
Voice of the Customer
Method development strategies
1 Method Development
Outputs
A fit for purpose method
2 Risk Assessment
Identify the design space
Inputs
Parameters selected for study
Parameters which can be fixed
Inputs
Noise Factors selected for study
3 Method Robustness
4 Method Ruggedness
5 Control Definition
6 Method Validation and use
7 Continuous Improvement and Change Control
Outputs
Learnings
Design Intent
Design Selection
EMPACT
Control Definition
Control Verification

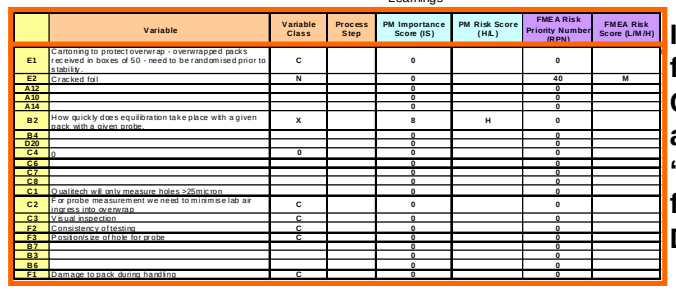
Variable
Variable Class
Process Step
PM Importance Score (IS)
PM Risk Score (H/L)
FMEA Risk Priority Number (RPN)
FMEA Risk Score (L/M/H)

Inclusion of factors to be Controlled alongside ''X'' and ''N'' factors for a DPI product

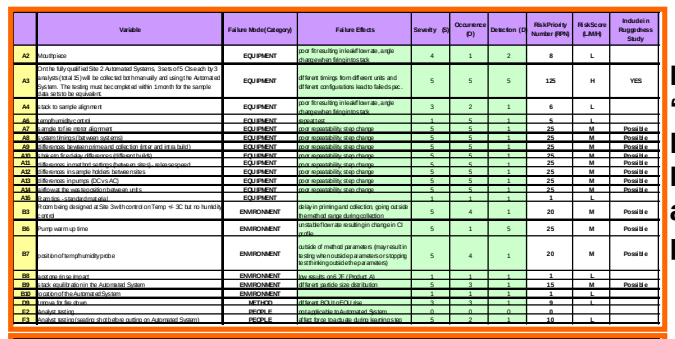
Variable
Failure Mode (Category)
Failure Effects
Severity (S)
Occurrence (O)
Detection (D)
Risk Priority Number (RPN)
Risk Score (L/M/H)
Include in Ruggedness Study

Example of ''N'' factors held in EMPACT for an MDI product

	Variable	Accuracy	Precision	Repeatability	Reproducibility	Selectivity	Linearity	Importance Score (IS)	Risk Score (H/L)	Include in Method Development (YES)	Include in Robustness Study
A5	Instrument vibration	5	5	5	5	1	1	22	H		YES
A15	Damping system different from site 1 to site 2	1	3	3	3	1	1	12	H		YES
B1	lock at impact on time between priming and collection	5	5	5	5	1	1	22	H		YES
D1	Shake duration	5	5	5	5	1	1	22	H	YES	YES
D2	Shake Speed	5	5	5	5	1	1	22	H		YES
D3	Hold time	5	5	5	5	1	1	22	H	YES	YES
D4	Speed to actuate	5	5	5	5	1	1	22	H		YES
D8	Release speed	7	5	5	5	1	1	24	H		YES

Example of ''X'' factors held in EMPACT for an MDI product

Outputs from Prioritisation matrices and FMEA exercises that come out of AM-QbD Risk Assessments held in EMPACT templates.

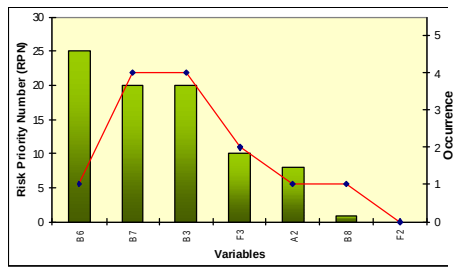
Risk Priority Number (RPN)
Occurrence
Variables
B6
B7
B3
F3
A2
B8
F2

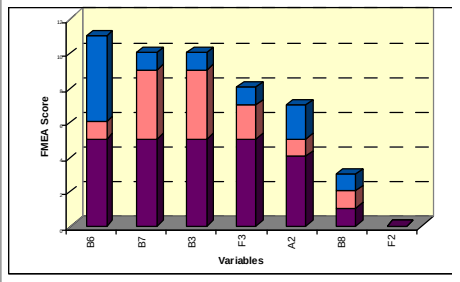
FMEA Score
Variables
B6
B7
B3
F3
A2
B8
F2

Any proposed changes that take the method outside its proven design space are risk assessed and for high risk changes an evaluation or equivalence exercise is performed to assure method performance criteria are still met. Method specific learnings are used to update the method knowledge repository. Technique learnings are used to enhance the risk assessment process for future methods. This is particularly important for automated methods for OINDPs.

QbD for Inhaled Product Automation

• Cross inhaled Project application (automation MDI and DPI)

• QbD example for introduction of inhaled product automation has been presented to FDA

• Reduction of potential Analyst error using Automation

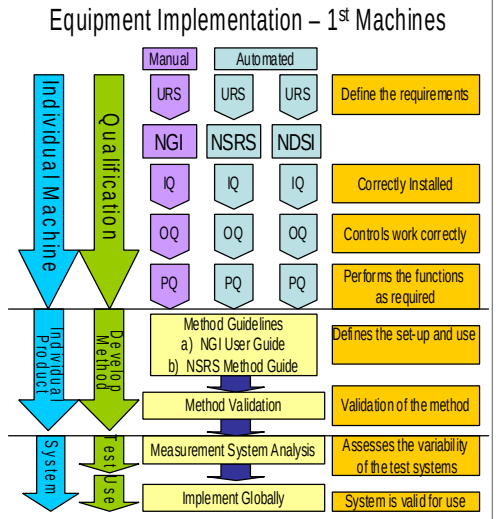
Equipment Implementation – 1st Machines
Individual Machine
Qualification
Manual
Automated
URS
URS
URS
Define the requirements
NGI
NSRS
NDSI
IQ
IQ
IQ
Correctly Installed
OQ
OQ
OQ
Controls work correctly
PQ
PQ
PQ
Performs the functions as required
Individual Product
Develop Method
Method Guidelines
a) NGI User Guide
b) NSRS Method Guide
Defines the set-up and use
Method Validation
Validation of the method
System
Test Use
Measurement System Analysis
Assesses the variability of the test systems
Implement Globally
System is valid for use

Semi-Automated NGI DPI Sample Solution Preparation

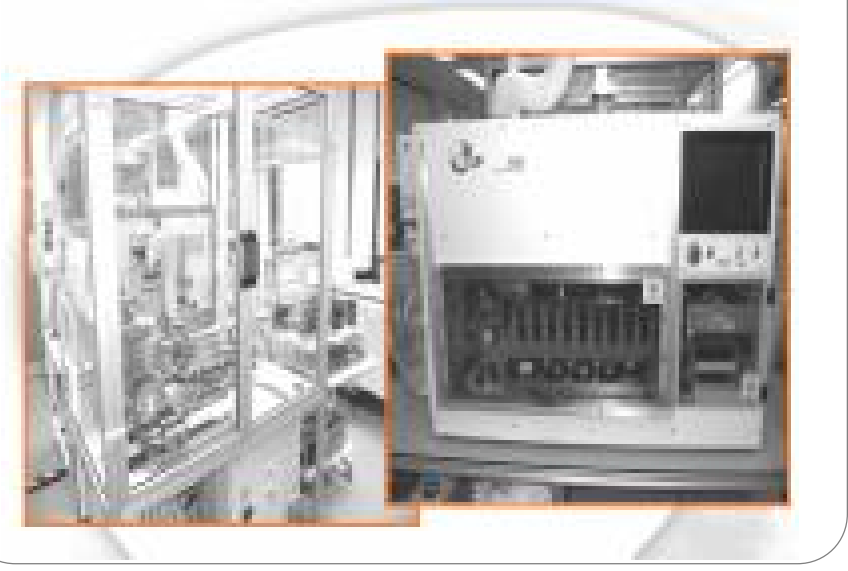

Method Performance Criteria

Method Performance Criteria are driven by an understanding of the process monitoring and control requirements and the ability to maintain the Product Design Space, i.e. the ability to meet the required critical quality attributes (CQAs). CQAs are identified, through a thorough understanding of those attributes of a drug substance or a drug product which may need to be controlled in order to assure the safety or efficacy of a product. This control can be achieved via specification limits, In Process Controls, in line monitoring and Process Analytical Technology etc. Method Performance Criteria are defined by using a rigorous approach for identifying all the potential method factors that need to be controlled to assure method performance and through the use of risk assessment tools and prioritised experimentation that eliminate areas of risk.

Blister Strip Apparatus (BSA)

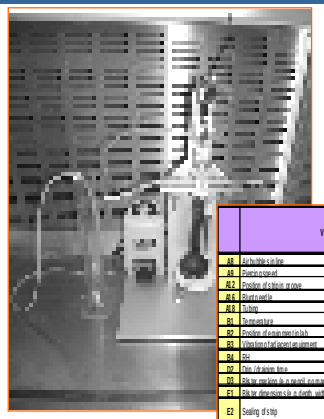

- Manual manipulation of the strip
- Controlled alignment of piercing
- Standardised process
- Sample recovery by pumped solvent
- Volumetric process
- Removal of wash bottles

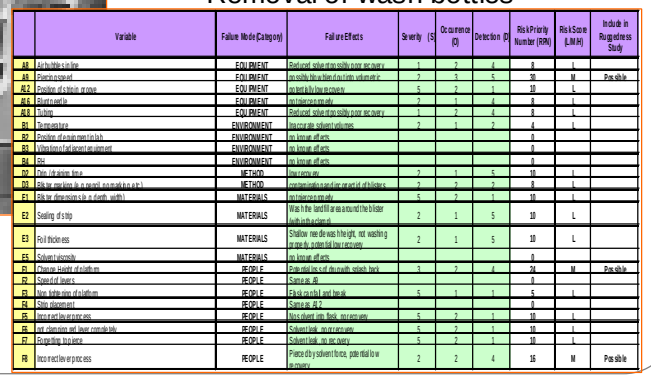
Variable
Failure Mode (Category)
Failure Effects
Severity (S)
Occurrence (O)
Detection (D)
Risk Priority Number (RPN)
Risk Score (L/M/H)
Include in Ruggedness Study

Conclusion

As demonstrated, the AM-QbD Risk Assessment process for OINDPs has been fully considered to fully mitigate the analytical risk associated with testing OINDPs in different laboratories within GSK. Key to this process is the scoring system (for prioritising potential method factors that need to be mitigated) that has been developed, piloted and established within GSK R&D.

Corresponding author: Andrew J Crumpton. World Wide Inhaled Product Development, GSK, R&D

Created by Information Design